ABSTRACT 317

Background: While twice-daily treatment (bid) with carbamazepine extended-release capsules (CBZ-ERC) (Equetro™; Shire, Wayne, Pa) has been shown to provide significant therapeutic benefit to patients with acute bipolar mania, retrospective findings suggest that once-daily dosing of CBZ-ERC may be similarly beneficial. Thus, the current study was undertaken to compare prospectively once-nightly (qhs) CBZ-ERC dosing with bid dosing in patients with bipolar disorder.

Methods: All participants in the current 12-week, double-blind trial were adult outpatients experiencing either an acute manic or mixed bipolar episode at study entry. At baseline, study participants were randomized to treatment with either bid or qhs CBZ-ERC. In both treatment groups, patients received a total CBZ-ERC dose of 200 to 1600 mg/d, with optimal doses determined via dose titration over the first 4 weeks postbaseline. Safety and tolerability were assessed at weeks 1, 2, 3, 4, 6, 8, and 12.

Results: Treatment with CBZ-ERC was relatively safe and well tolerated in both treatment groups, with most treatment-related adverse events mild or moderate. Among the most common adverse events (≥10% incidence) in both treatment arms (bid, n = 53; qhs, n = 58) were nausea (bid, 30.2%; qhs, 24.1%), dizziness (bid, 24.5%; qhs, 17.2%), headache (bid, 22.6%; qhs, 24.1%), sedation (bid, 15.1%; qhs, 12.1%), and blurred vision (bid, 9.4%; qhs, 10.3%). Fatigue (bid, 17.0%; qhs, 6.9%), somnolence (bid, 15.1%; qhs, 8.6%), and increased appetite (bid, 11.3%; qhs, 3.4%) were more common among patients treated bid than among patients treated qhs, whereas rates of emesis (bid, 3.8%; qhs, 12.1%) and dry mouth (bid, 11.3%; qhs, 17.2%) were higher in the qhs arm than in the bid arm. No serious treatment-related rashes, blood dyscrasias, or cardiac abnormalities were reported. With regard to metabolic adverse effects, 0% and 3.4% of bid- and qhs-treated patients, respectively, reported weight gain, while mean nonfasting serum total cholesterol levels increased in both treatment arms between baseline and end point (bid, +18.1 mg/dL; qhs, +12.5 mg/dL).

Conclusions: Carbamazepine extended-release capsules, whether administered using a bid or a qhs dosing schedule, were safe and well tolerated among patients experiencing acute manic or mixed bipolar episodes. Furthermore, metabolic parameters were generally not dramatically affected by CBZ-ERC therapy.

A RANDOMIZED, DOUBLE-BLIND, PARALLEL-GROUP TRIAL OF TWICE-DAILY AND ONCE-DAILY CARBAMAZEPINE EXTENDED-RELEASE CAPSULES IN BIPOLAR DISORDER: ANALYSIS OF SAFETY AND TOLERABILITY

Lawrence Ginsberg, MD, Red Oak Psychiatry Associates, Houston, Texas; **Richard H. Weisler, MD,** Duke University Medical Center, Durham, North Carolina, and the University of North Carolina, Chapel Hill, North Carolina; **Thomas Gazda, MD,** St Luke's Medical Center, Scottsdale, Arizona; **Joseph Kerkering, MBA,** Shire, Wayne, Pennsylvania

BACKGROUND

- Although carbamazepine (CBZ) has been used in the treatment of bipolar disorder for more than 3 decades,[1,2] CBZ extended-release capsules (CBZ-ERC) (Equetro™; Shire, Wayne, Pa) are the first CBZ formulation to be approved by the US Food and Drug Administration for this indication
- Phase 3 clinical trials examining the use of CBZ-ERC to treat patients experiencing acute manic or mixed bipolar episodes have established that this agent is safe and effective when administered according to a twice-daily (bid) dosing schedule[3,4]
- Findings made in a recent retrospective chart study suggest that once-daily dosing of CBZ-ERC may be comparable to bid dosing with regard to efficacy and safety[5]
- To gain a more complete understanding of the relative efficacy and safety of once-per-day CBZ-ERC dosing, a 12-week, randomized, double-blind, placebo-controlled trial prospectively comparing a once-nightly (qhs) CBZ-ERC treatment regimen with the standard bid CBZ-ERC treatment regimen in patients with Bipolar I Disorder was conducted
 - The current outpatient trial employed a 4-week CBZ-ERC dose titration schedule, in contrast to the more rapid 1-week titration schedule followed in previous CBZ-ERC trials, which involved inpatients with acute bipolar symptoms

METHODS

- Adult outpatients with Bipolar I Disorder (Young Mania Rating Scale score ≥16), most recent episode manic or mixed, were recruited from among those who presented to any of 12 participating US study sites between January and October 2005
- Recruited patients were assessed for study eligibility over a 2- to 10-day screening period, after which those who were found to meet the necessary eligibility criteria were randomly assigned to receive either qhs or bid treatment with CBZ-ERC (Figure 1)
 - Patients in the qhs treatment arm received their entire daily CBZ-ERC dose in the evening
 - Patients in the bid treatment arm received half of their total daily CBZ-ERC dose in the morning and the remaining half in the evening
- Following the initiation of CBZ-ERC therapy at a total daily dose of 200 mg, treatment doses were adjusted as needed over the course of a 4-week dose titration period and then held constant for the remainder of the study. During the dose titration period, total daily doses were increased or decreased in 200-mg steps once every 3 to 4 days as necessary for efficacy or tolerability (allowed daily dose range, 200-1600 mg; target daily dose, 800 mg)
- Adverse event recording and vital sign evaluation were performed weekly during the 4-week dose titration period and then at 6, 8, and 12 weeks from baseline. In addition, electrocardiograms were obtained at 4 and 12 weeks from baseline, while laboratory parameters were assessed at 4, 8, and 12 weeks from baseline
 - For patients who withdrew from the study before its completion, all scheduled week 12 safety evaluations were performed on the final day of study treatment or as soon as possible (≤2 days) thereafter
- Each patient underwent an additional follow-up evaluation of treatment safety and tolerability (via telephone or in-person interview) 30 days after receiving his or her final dose of study drug

RESULTS

Patient Characteristics, Patient Disposition, and Treatment Dosing

- The safety population in the current trial comprised 111 patients; the qhs (n = 58) and bid (n = 53) treatment arms were similar with regard to demographics and baseline disease characteristics (Table 1)
- A total of 47 patients (42.3%) completed the entire course of study treatment. Reasons for early termination included loss to follow-up (n = 23 [20.7%]), adverse events/serious adverse events (n = 16 [14.4%]), and withdrawal of consent (n = 15 [13.5%]), among others (Table 2)
- A total of 31 patients (27.9%) used concomitant psychotropic medications during the course of study treatment
- More than half of all patients in both treatment arms were exposed to CBZ-ERC for greater than 4 weeks during the trial (qhs arm, 62.1%; bid arm, 67.9%). However, patients in the qhs treatment arm had a somewhat shorter mean duration of exposure to study drug than did patients in the bid treatment arm (6.91 weeks vs 8.18 weeks)
- The mean total daily CBZ-ERC dose for qhs-treated patients was 656 mg, while the corresponding mean daily dose for bid-treated patients was 727 mg

Figure 1. Study Design

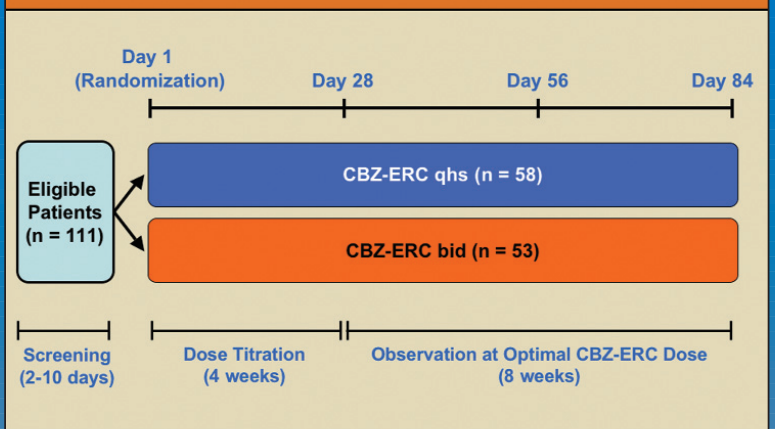
Day 1
(Randomization)
Day 28
Day 56
Day 84
Eligible Patients (n = 111)
CBZ-ERC qhs (n = 58)
CBZ-ERC bid (n = 53)
Screening
(2-10 days)
Dose Titration
(4 weeks)
Observation at Optimal CBZ-ERC Dose
(8 weeks)

qhs = once nightly; bid = twice daily; CBZ-ERC = carbamazepine extended-release capsules.

Figure 2. Dizziness and Somnolence in Clinical Trials of CBZ-ERC Therapy for Manic or Mixed Bipolar Symptoms*

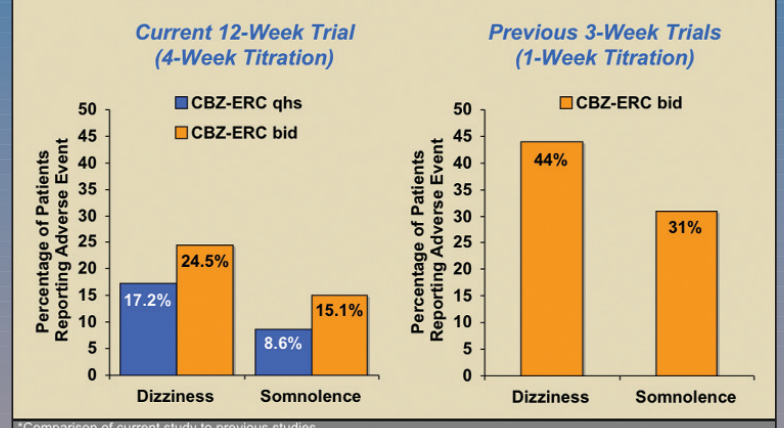
Current 12-Week Trial
(4-Week Titration)
Previous 3-Week Trials
(1-Week Titration)
CBZ-ERC qhs
CBZ-ERC bid
Percentage of Patients Reporting Adverse Event
50
45
40
35
30
25
20
15
10
5
0
17.2%
24.5%
8.6%
15.1%
44%
31%
Dizziness
Somnolence

*Comparison of current study to previous studies.
CBZ-ERC = carbamazepine extended-release capsules; qhs = once nightly; bid = twice daily.
Source: Weisler RH, et al. Presented at: US Psychiatric and Mental Health Congress; November 18-21, 2004; San Diego, Calif.

Figure 3. Severity of Treatment-Emergent Adverse Events

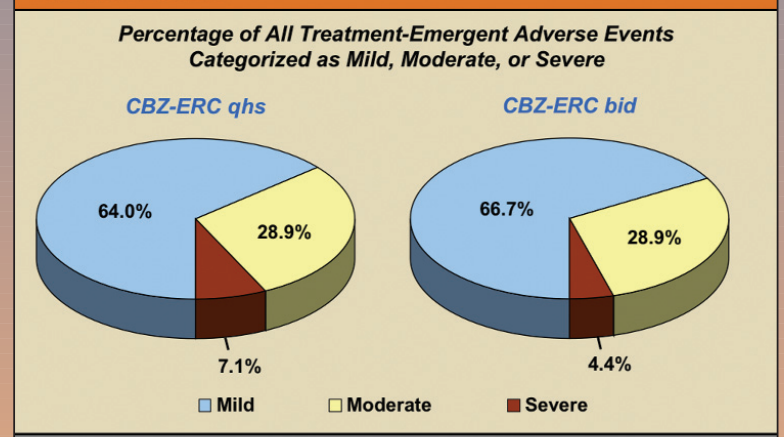
Percentage of All Treatment-Emergent Adverse Events Categorized as Mild, Moderate, or Severe
CBZ-ERC qhs
CBZ-ERC bid
64.0%
28.9%
7.1%
66.7%
28.9%
4.4%
Mild
Moderate
Severe

qhs = once nightly; bid = twice daily; CBZ-ERC = carbamazepine extended-release capsules.

Table 1. Baseline Patient Characteristics

Characteristic	Treated qhs	Treated bid
Enrolled, N	58	53
Gender, n (%)		
Male	19 (32.8%)	23 (43.4%)
Female	39 (67.2%)	30 (56.6%)
Race/Ethnicity, n (%)		
White	44 (75.9%)	41 (77.4%)
Black	8 (13.8%)	6 (11.3%)
Hispanic	6 (10.3%)	6 (11.3%)
Mean weight (lb) ± SD	191.8 ± 52.0	178.1 ± 49.0
Mean YMRS total score ± SD	20.6 ± 6.6	22.8 ± 7.3
Mean HDRS total score ± SD	13.7 ± 6.9	13.6 ± 7.2

qhs = once nightly; bid = twice daily; YMRS = Young Mania Rating Scale; SD = standard deviation; HDRS = Hamilton Depression Rating Scale.

Table 2. Patient Disposition

	Treated qhs (%)	Treated bid (%)
Enrolled, N	58	53
Completed study	21 (36.2%)	26 (49.1%)
Withdrew before completion	37 (63.8%)	27 (50.9%)
Lost to follow-up	10 (17.2%)	13 (24.5%)
Withdrew due to adverse event	10 (17.2%)	6 (11.3%)
Withdrew consent	10 (17.2%)	5 (9.1%)
Withdrew due to lack of efficacy	3 (5.2%)	1 (1.9%)
Died	0	1* (1.9%)
Other	4 (6.9%)	1 (1.9%)

qhs = once nightly; bid = twice daily.
*Patient died of an accidental methadone overdose unrelated to study treatment.

Table 3. Summary of Common Treatment-Emergent Adverse Events*

Adverse Event	Treated qhs, n (%)	Treated bid, n (%)
Nausea	14 (24.1%)	16 (30.2%)
Dizziness	10 (17.2%)	13 (24.5%)
Headache	14 (24.1%)	12 (22.6%)
Dry mouth	10 (17.2%)	6 (11.3%)
Fatigue	4 (6.9%)	9 (17.0%)
Sedation	7 (12.1%)	8 (15.1%)
Somnolence	5 (8.6%)	8 (15.1%)
Emesis	7 (12.1%)	2 (3.8%)
Increased appetite	2 (3.4%)	6 (11.3%)
Blurred vision	6 (10.3%)	5 (9.4%)

qhs = once nightly; bid = twice daily.
*Common adverse events were those that occurred in ≥10% of patients in 1 or both treatment arms.

REFERENCES

1. Takezaki H, Hanaoka M. The use of carbamazepine (Tegretol) in the control of manic depressive psychosis and other manic depressive states. *Seishin Igaku.* 1971;13:173-183.
2. Okuma T, Kishimoto A, Inoue K, Matsumoto H, Ogura A. Anti-manic and prophylactic effects of carbamazepine (Tegretol) on manic depressive psychosis. A preliminary report. *Folia Psychiatr Neurol Jpn.* 1973;27:283-297.
3. Weisler RH, Kalali AH, Ketter TA. A multicenter, randomized, double-blind, placebo-controlled trial of extended-release carbamazepine capsules as monotherapy for bipolar disorder patients with manic or mixed episodes. *J Clin Psychiatry.* 2004;65:478-484.
4. Weisler RH, Keck PE Jr, Swann AC, Cutler AJ, Ketter TA, Kalali AH. Extended-release carbamazepine capsules as monotherapy for acute mania in bipolar disorder: a multicenter, randomized, double-blind, placebo-controlled trial. *J Clin Psychiatry.* 2005;66:323-330.
5. Ginsberg LD. Safety and efficacy of carbamazepine extended-release capsules in patients with bipolar disorder: QD vs BID. *Ann Clin Psychiatry.* 2006;18(suppl 1):27-30.
6. Weisler RH, Hirschfeld R, Cutler AJ, et al. Safety and tolerability of extended-release carbamazepine in bipolar disorder: results of 2 pooled clinical trials. Presented at: US Psychiatric and Mental Health Congress; November 18-21, 2004; San Diego, Calif.

RESULTS (CONT)

Treatment Safety

- Treatment-emergent adverse events were reported by 86.2% of patients in the qhs treatment arm and by 90.6% of patients in the bid treatment arm. Incidence rates of commonly occurring adverse events are summarized in Table 3
 - A number of adverse events arose more commonly in association with bid dosing than with qhs dosing, including nausea, dizziness, fatigue, somnolence, and increased appetite. In contrast, dry mouth and emesis were observed more commonly in the qhs treatment arm than in the bid treatment arm
 - Rates of dizziness and somnolence for patients in both treatment arms were lower than the corresponding rates for CBZ-ERC–treated patients in previous short-term randomized trials, which involved inpatients with acute manic or mixed bipolar symptoms and thus employed a more rapid 1-week dose titration schedule[6] (Figure 2)
- The large majority of treatment-emergent adverse events in both treatment arms were of mild to moderate severity (Figure 3)
 - Adverse events judged to be severe and to have a possible or probable relation to study drug were reported by 5 patients in the qhs treatment arm (night sweats [n = 1]; dizziness and headache [n = 1]; hypersensitivity [n = 1]; fatigue [n = 1]; increased weight, increased appetite, and syncope [n = 1]) and in 1 patient in the bid treatment arm (headache)
- Rates of adverse event–related treatment discontinuation did not differ significantly between the qhs and bid treatment arms (qhs arm, 17.2% vs bid arm, 11.3%; *P*=.427)
- Serious adverse events occurred in 4 qhs-treated patients (worsening bipolar disorder [n = 1], allergic reaction [n = 1], syncope [n = 1], traffic accident and resulting spinal pain [n = 1]) and in 2 bid-treated patients (accidental methadone overdose resulting in death [n = 1] and worsening bipolar disorder [n = 1]). Of these serious adverse events, 2 events (allergic reaction [n = 1] and syncope [n = 1], both in the qhs treatment arm) were judged to have a possible or probable association with the study drug
- The qhs and bid dosing schedules were both associated with minimal mean changes in body weight between baseline and final evaluation (qhs arm, +1.1 kg; bid arm, +1.7 kg)
 - Two patients in the qhs treatment arm (3.4%) reported adverse occurrences of weight gain during the course of CBZ-ERC therapy
- Relative to bid-treated patients, qhs-treated patients experienced larger mean changes in nonfasting blood glucose levels (qhs arm, +14.6 mg/dL; bid arm, +1.8 mg/dL) and nonfasting blood cholesterol levels (qhs arm, +18.1 mg/dL; bid arm, +12.5 mg/dL) between baseline and final evaluation
 - Treatment-emergent hyperglycemia was documented in 2 patients in the qhs treatment arm (3.4%), while 1 patient in the bid treatment arm experienced a clinically significant increase in blood cholesterol levels (1.9%) during CBZ-ERC therapy
- No cases of aplastic anemia, agranulocytosis, or serious rash were documented in either treatment arm, and no serious electrocardiographic or vital sign abnormalities were seen in association with the study drug

CONCLUSIONS

- Carbamazepine extended-release capsules are generally safe and well tolerated when administered according to a qhs or a bid schedule for the treatment of patients with manic or mixed bipolar symptoms
 - Adverse events seen in association with both dosing schedules were, in the large majority of cases, mild to moderate in severity, and serious treatment-emergent adverse events showing a possible relation to CBZ-ERC therapy were uncommon
- Once-nightly dosing of CBZ-ERC may be somewhat better tolerated than bid dosing, given that a number of adverse events, including nausea, dizziness, somnolence, and increased appetite, occurred more frequently in the bid treatment arm than in the qhs treatment arm
- Both dosing schedules examined in the current study were associated with minimal weight gain. However, qhs-treated patients experienced numerically larger mean changes in blood glucose and total cholesterol levels than did bid-treated patients
 - The ability to interpret data regarding blood glucose and total cholesterol levels in the current study is limited, given that these parameters were assessed in nonfasting blood samples
- Patients in the current trial were less likely than patients in previous short-term trials who received CBZ-ERC for the treatment of manic or mixed bipolar symptoms to experience dizziness or somnolence, possibly due to the slower titration schedule used in the current trial
- Taken together with efficacy data from the current study, the data presented here indicate that qhs dosing may be an acceptable alternative to bid dosing of CBZ-ERC for patients with manic or mixed bipolar symptoms

Equetro is a trademark of Shire LLC.

Supported by funding from Shire.

Presented at the 46th Annual Meeting of the New Clinical Drug Evaluation Unit; June 12-15, 2006; Boca Raton, Florida.