SHR CRL i40368 peds 376 4/19/04 6:00 PM Page 1

NR376

ABSTRACT

Objective: To assess the effectiveness and safety of extended-release carbamazepine capsules (ERC-CBZ; SPD417) in the treatment of pediatric patients with bipolar disorder.

Method: A chart review of 79 children and adolescents aged 7–17 with *DSM-IV* bipolar disorder and treated with ERC-CBZ was conducted (mean age 12.8 ± 3.0 years; 39% female; 40.5% bipolar I, 29.1% bipolar II, 30.4% bipolar not otherwise specified). Charts of subjects who received ERC-CBZ in a private practice setting (LDG, Red Oak Psychiatry Associates, Houston, Tex) between February 1999 and July 2003 were reviewed. Treatment response was assessed with the Clinical Global Impressions–Improvement (CGI-I) scale (1 = marked improvement; 2 = moderate improvement). Relapse was defined as a mood change that occurs 4 weeks after initiation of treatment medication or the return of symptoms from the original episode.

Results: Forty-two subjects (53.2%) taking ERC-CBZ had marked to moderate improvement (CGI-I score: 1, 32.9%; 2, 20.3%). No subjects experienced moderate to marked worsening. Twenty-two patients (28%) relapsed during ERC-CBZ treatment (mean time to relapse = 166 days). Mixed symptoms were the most common bipolar illness presentation. The mean ERC-CBZ dose was 620.5 ± 199.5 mg/d, and the mean serum concentration was 7.1 ± 1.8 μg/mL. Dizziness (10.1%) and nausea (8.9%) were the most frequently reported side effects.

Conclusion: Extended-release carbamazepine appears effective in the treatment of pediatric patients with bipolar disorder and was well tolerated.

NOTE: Study protocol has changed since abstract submission.

[The following information concerns a use that has not been approved by the U.S. Food and Drug Administration.]

SAFETY AND EFFICACY OF EXTENDED-RELEASE CARBAMAZEPINE FOR PEDIATRIC BIPOLAR DISORDER PATIENTS

Lawrence D. Ginsberg, MD, Red Oak Psychiatry Associates, Houston, Texas

INTRODUCTION

- There is a paucity of studies on the treatment of pediatric bipolar disorder, with most pharmacological treatment guidelines relying upon open studies and case reports.[1] Many pediatric studies suggest the effective use of a mood stabilizer either as monotherapy or in combination with other treatments.[2-4] In a small, uncontrolled study of carbamazepine (CBZ), 9 of 11 pediatric patients responded to therapy with only mild adverse events[5]
- Many of the treatment options in pediatric bipolar disorder have been extrapolated from results in more extensive studies of adult bipolar disorder. Carbamazepine has been studied in adult bipolar disorder and has consistently demonstrated efficacy and tolerability comparable to lithium, with a pooled response rate of 52%.[6] However, these studies had limitations such as small patient populations or concomitant treatment with other medications, and all used immediate-release carbamazepine (IR-CBZ)
- Problems associated with IR-CBZ have been numerous adverse events, and the fact that it can require TID or QID dosing[7,8]
- Extended-release (ER) formulations of CBZ have been developed in recent years to decrease daily fluctuations in serum CBZ concentration,[9] improve dosing convenience, and decrease central nervous system side effects such as sedation, diplopia, confusion, and ataxia[10]
- A novel ER formulation—beaded, extended-release CBZ capsules (ERC-CBZ; SPD417)—is filled with 3 different types of beads: 25% immediate-release, 40% ER, and 35% enteric release
- Recently, 2 large, 3-week, multicenter, randomized, double-blind, placebo-controlled trials showed ERC-CBZ monotherapy to be effective in the treatment of manic symptoms in adult patients with bipolar disorder.[11,12] Based on the results of these 2 studies, a New Drug Application has been filed with the US Food and Drug Administration for an indication for ERC-CBZ in the treatment of bipolar disorder
- Little is known about the long-term efficacy and tolerability of ERC-CBZ in the treatment of pediatric bipolar disorder. The purpose of this study is to evaluate the effectiveness, safety, and tolerability of ERC-CBZ in pediatric patients with bipolar disorder

METHODS

- This is an interim report of an ongoing retrospective chart review
- Records of pediatric patients (age ≤17) with a *DSM-IV* diagnosis of bipolar disorder who were treated with ERC-CBZ at a private practice (LDG, Red Oak Psychiatry Associates, Houston, Tex) between October 1998 and February 2004 (n=202) were included in this study
- Charts were reviewed for relapse, adverse events, scores on the Clinical Global Impression–Improvement (CGI-I) and Clinical Global Impression–Severity (CGI-S) scales, CBZ blood levels, and white blood cell count
- Patients were considered to have responded to therapy if they achieved a CGI-I score ≤3. Patients were considered to have relapsed if they had a CGI-I ≥4 after a response had been previously observed

Figure 1. Patient Population

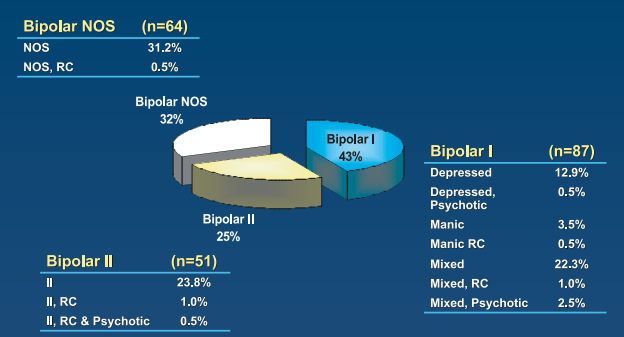

Bipolar NOS (n=64)
NOS 31.2%
NOS, RC 0.5%
Bipolar NOS 32%
Bipolar I 43%
Bipolar II 25%
Bipolar I (n=87)
Depressed 12.9%
Depressed, Psychotic 0.5%
Manic 3.5%
Manic RC 0.5%
Mixed 22.3%
Mixed, RC 1.0%
Mixed, Psychotic 2.5%
Bipolar II (n=51)
II 23.8%
II, RC 1.0%
II, RC & Psychotic 0.5%

Data on file, Lawrence D. Ginsberg, MD, Red Oak Psychiatry Associates.
RC = rapid cycling; NOS = not otherwise specified

Figure 2. Severity of Illness on the CGI-S Scale at ERC-CBZ initiation

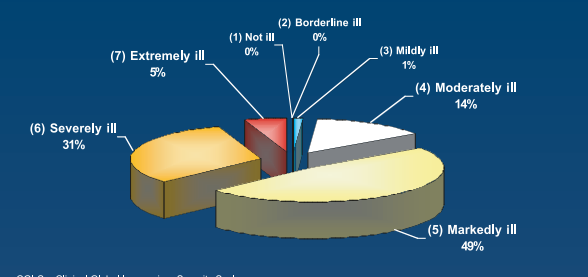

(1) Not ill 0%
(2) Borderline ill 0%
(3) Mildly ill 1%
(4) Moderately ill 14%
(5) Markedly ill 49%
(6) Severely ill 31%
(7) Extremely ill 5%

CGI-S = Clinical Global Impression–Severity Scale

Figure 3. Response and Relapse Rates of Pediatric Bipolar Disorder Patients on ERC-CBZ

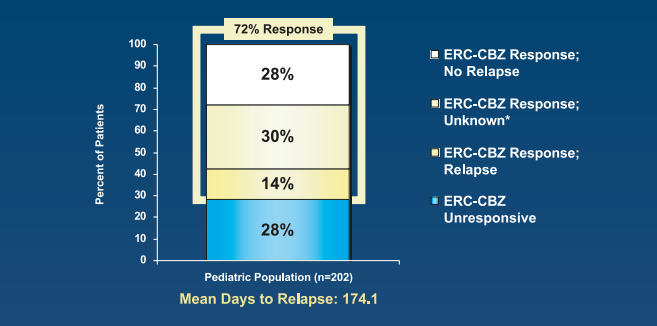

72% Response
28%
30%
14%
28%
Percent of Patients
Pediatric Population (n=202)
Mean Days to Relapse: 174.1
ERC-CBZ Response; No Relapse
ERC-CBZ Response; Unknown*
ERC-CBZ Response; Relapse
ERC-CBZ Unresponsive

ERC-CBZ response is defined as achieving a CGI-I ≤ 3. Relapse is defined by a change in CGI-I to ≥ 4 after an observed ERC-CBZ response.
*Response unknown includes: lost to follow-up, moved out of location; switched physicians, couldn't afford treatment, etc.
Data on file, Lawrence D. Ginsberg, MD, Red Oak Psychiatry Associates.

Table 1. Patient Demographics and ERC-CBZ Results

Pediatric Patients	202
Gender (% Female)	37.6%
Mean age (y ± SD)	12.5 ± 3.2
Age Range (y)	4 – 17
Mean ERC-CBZ dose* (mg/d)	594.5 ± 231.5
Mean ERC-CBZ blood level* (μg/mL)	7.1 ± 1.8
ERC-CBZ Monotherapy at initiation	29.7%
Patients dosed QD on ERC-CBZ	5.4%

CGI-I = Clinical Global Impression–Improvement Scale
*Mean dose and blood levels are at patient's best CGI-I score.
Data on file, Lawrence D. Ginsberg, MD, Red Oak Psychiatry Associates.

Figure 4. Analysis of ERC-CBZ Responders' CGI-I Scores

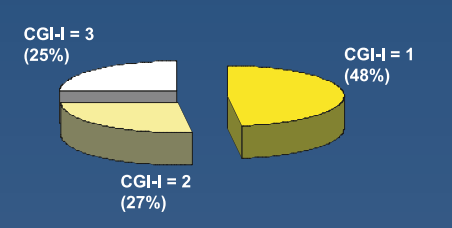

CGI-I = 3 (25%)
CGI-I = 1 (48%)
CGI-I = 2 (27%)

CGI-I = Clinical Global Impression – Improvement scale
ERC-CBZ = Extended-release carbamazepine capsules

Figure 5. Dose Distribution of ERC-CBZ at Best CGI-I

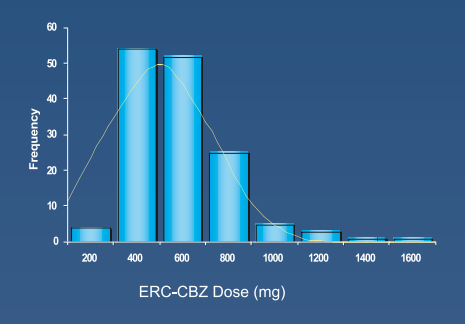

Frequency
200
400
600
800
1000
1200
1400
1600
ERC-CBZ Dose (mg)

Table 2. Treatment-Emergent Adverse Events on ERC-CBZ

Event	(n=202)
Somnolence	11.9%
Nausea	6.9%
Dizziness	6.9%
Headaches	5.0%
Low WBC	3.5%
Vomiting	3.5%
Rash	3.0%
Dyspepsia	2.0%

WBC = white blood cell count
Adverse events reported with an incidence ≥ 2%.
Data on file, Lawrence D. Ginsberg, MD, Red Oak Psychiatry Associates.

ERC-CBZ = extended-release carbamazepine capsules

RESULTS

- The mean age of the 202 pediatric patients in this study was 12.5 years. At ERC-CBZ initiation, ages ranged from 4–17, and 38% of patients were female. Thirty percent of patients were initiated on ERC-CBZ as monotherapy, while 5.4% of patients were maintained on a once-daily dose of ERC-CBZ **(Table 1)**
- Forty-four percent of patients were diagnosed with bipolar I disorder, 25% with bipolar II disorder, and 31% had a diagnosis of bipolar disorder not otherwise specified **(Figure 1)**
- The severity of bipolar illness at ERC-CBZ initiation was determined using the CGI-S scale. Eighty-five percent of patients were determined to be at least markedly ill, scoring ≥ 5 on the CGI-S **(Figure 2)**
- One hundred forty-five (72%) patients responded to ERC-CBZ
- Of the responders, 28% did not relapse, 30% were lost to follow-up, and 14% relapsed. The average time to relapse was 174 days (95% CI, 100–248 days) **(Figure 3)**
- Analysis of ERC-CBZ responders revealed 48% of patients having achieved a CGI-I score of 1, 27% a score of 2, and 25% a score of 3 **(Figure 4)**
- At patients' best CGI-I score, the mean ERC-CBZ dose was 595 ± 232 mg/d, and the average ERC-CBZ blood level was 7.1 ± 1.8 μg/mL **(Table 1; Figure 5)**
- Common treatment-emergent adverse events included somnolence (11.9%), nausea (6.9%), and dizziness (6.9%) **(Table 2)**
- There was no evidence of agranulocytosis or aplastic anemia

CONCLUSIONS

- This large retrospective study demonstrates that ERC-CBZ is effective for the treatment of pediatric bipolar disorder
- ERC-CBZ was effective in a broad spectrum of bipolar disorder in the pediatric population
- There was a low incidence of side effects, with no evidence of agranulocytosis or aplastic anemia

REFERENCES

1. Practice guideline for the treatment of patients with bipolar disorder (revision). *Am J Psychiatry*. 2002;159:1-50.
2. Biederman J, Mick E, Bostic JQ, et al. The naturalistic course of pharmacologic treatment of children with maniclike symptoms: a systemic chart review. *J Clin Psychiatry*. 1998;59:628-637.
3. Kowatch RA, Suppes T, Carmody TJ, et al. Effect size of lithium, divalproex sodium, and carbamazepine in children and adolescents with bipolar disorder. *J Am Acad Child Adolesc Psychiatry.* 2000;39:713-720.
4. Kowatch RA, Sethuraman G, Hume JH, et al. Combination pharmacotherapy in children and adolescents with bipolar disorder. *Biol Psychiatry.* 2003;53:978-984.
5. Bouvard MP, Bayle F, Dugas M. Open trial of carbamazepine in the prevention of recurrence of bipolar disorder in adolescents. *Encephale.* 1993;19:591-600.
6. McElroy SL, Keck PE Jr. Pharmacologic agents for the treatment of acute bipolar mania. *Biol Psychiatry.* 2000;48:539-557.
7. Jensen PK, Moller A, Gram L, et al. Pharmacokinetic comparison of two carbamazepine slow-release formulations. *Acta Neurol Scand.* 1990;82:135-137.
8. Hoppener RJ, Kuyer A, Meijer JW, et al. Correlation between daily fluctuations of carbamazepine serum levels and intermittent side effects. *Epilepsia.* 1980;21:341-350.
9. Canger R, Altamura AC, Belvedere O, et al. Conventional vs controlled-release carbamazepine: a multicenter, double- blind, cross-over study. *Acta Neurol Scand.* 1990;82:9-13.
10. Mirza WU, Rak IW, Thadani VM, et al. Six-month evaluation of Carbatrol (extended-release carbamazepine) in complex partial seizures. *Neurology.* 1998;51:1727-1729.
11. Weisler RH, Kalali AH, Ketter TA, and the SPD417 Study Group. A multicenter, randomized, double-blind, placebo-controlled trial of extended-release carbamazepine capsules as monotherapy for bipolar disorder patients with manic or mixed episodes. *J Clin Psychiatry.* In press.
12. Data on file, Shire Pharmaceutical Development. 2003.

Supported by funding from Shire.