SHR CRL i40368 Gins Adt 737 5.0 4/19/04 5:59 PM Page 1

NR737

ABSTRACT

Objective: To assess the effectiveness and safety of extended-release carbamazepine capsules (ERC-CBZ; SPD417) in the treatment of bipolar disorder.

Method: A chart review of 101 adult outpatients with *DSM-IV* bipolar disorder and treated with ERC-CBZ was conducted (mean age 32.8 ± 10.5 years; 71% female; 70% bipolar I, 18% bipolar II, 12% bipolar not otherwise specified). Charts of subjects who received ERC-CBZ in a private practice setting (LDG, Red Oak Psychiatry Associates, Houston, Tex) between October 1998 and August 2003 were reviewed. Treatment response was assessed with the Clinical Global Impression–Improvement (CGI-I) scale (1 = very marked improvement; 2 = moderate improvement). Relapse was defined as a mood change that occurs 4 weeks after initiation of medication or the return of symptoms from the original episode.

Results: Forty-four subjects (44%) taking ERC-CBZ had marked to moderate improvement (CGI-I score: 1, 23%; 2, 21%). No subjects experienced moderate to marked worsening. Twenty-six patients (26%) relapsed during ERC-CBZ treatment (mean time to relapse = 180 days). Mixed symptoms were the most common bipolar illness presentation. The mean ERC-CBZ dose was 666.1 ± 267.8 mg/d, and the mean serum concentration was 7.4 ± 2.3 μg/mL. Somnolence (9%) and nausea (8%) were the most frequently reported side effects.

Conclusion: Extended-release carbamazepine appears effective in the treatment of bipolar disorder and was well tolerated.

NOTE: Study protocol has changed since abstract submission.

[The following information concerns a use that has not been approved by the U.S. Food and Drug Administration.]

SAFETY AND EFFICACY OF EXTENDED-RELEASE CARBAMAZEPINE FOR ADULT BIPOLAR DISORDER PATIENTS

Lawrence D. Ginsberg, MD, Red Oak Psychiatry Associates, Houston, Texas

INTRODUCTION

- Carbamazepine (CBZ) has been studied in at least 14 double-blind, controlled trials, in which it consistently demonstrated efficacy in acute mania comparable to lithium, with a pooled response rate of 52%.[1] However, these studies had limitations such as small patient populations or concomitant treatment with other medications, and all used immediate-release CBZ (IR-CBZ)
- IR-CBZ has been associated with numerous adverse effects and can require TID or QID dosing[2,3]
- Extended-release (ER) formulations of CBZ have been developed in recent years to decrease daily fluctuations in serum CBZ concentration and improve dosing convenience. These formulations of CBZ have been associated with lower peak serum concentrations,[4] decreased circadian toxicity,[5] and decreased central nervous system side effects, such as sedation, diplopia, confusion, and ataxia[6]
- A novel ER formulation—beaded, extended-release CBZ capsules (ERC-CBZ; SPD417)—is filled with 3 different types of beads: 25% immediate release, 40% ER, and 35% enteric release
- ERC-CBZ monotherapy has been shown to be effective in the treatment of manic symptoms in patients with bipolar disorder in two 3-week, multicenter, randomized, double-blind, placebo-controlled trials.[7,8] Based on the results of these 2 studies, a New Drug Application has been filed with the US Food and Drug Administration for an indication for ERC-CBZ in the treatment of bipolar disorder
- The objectives of this study were to evaluate the effectiveness, safety, and tolerability of ERC-CBZ in adult patients with bipolar disorder

METHODS

- This is an interim report of an ongoing retrospective chart review
- Records of patients with a *DSM-IV* diagnosis of bipolar disorder who were treated with ERC-CBZ at a private practice (LDG, Red Oak Psychiatry Associates, Houston, Tex) between October 1998 and January 2004 (n=248) were included in this study
- Charts were reviewed for relapse, adverse events, weight, scores on the Clinical Global Impression–Improvement (CGI-I) and Clinical Global Impression–Severity (CGI-S) scales, CBZ blood levels, and white blood cell count
- Patients were considered to have responded to therapy if they achieved a CGI-I score of ≤ 3. Patients were considered to have relapsed if they had a CGI-I of ≥ 4 after a response had been previously observed

Supported by funding from Shire.

Table 1. Study Population

	Adult
Patients	248
Gender (% Female)	71%
Mean age (y ± SD)	34.7 ± 11.6
Age Range (y)	18 – 84
Mean ERC-CBZ dose* (mg/d)	617.0 ± 231.4
Mean ERC-CBZ blood level* (μg/mL)	7.1 ± 1.8
ERC-CBZ Monotherapy at initiation	49%
Patients dosed QD on ERC-CBZ	6%

CGI-I = Clinical Global Impression – Improvement scale
ERC-CBZ = Extended-release carbamazepine capsules
*Mean dose and blood levels are at patientís best CGI-I score.
Data on file, Lawrence D. Ginsberg, MD, Red Oak Psychiatry Associates.

Table 2. Treatment-Emergent Adverse Events on ERC-CBZ

Event	(n=248)
Somnolence	11.3%
Nausea	8.1%
Dizziness	7.7%
Rash	4.8%
Headaches	3.2%
Increased appetite	2.4%
Vomiting	2.0%
Blurred vision	2.0%
Weight gain	2.0%

Adverse events reported with an incidence ≥ 2%.
Data on file, Lawrence D. Ginsberg, MD, Red Oak Psychiatry Associates.

ERC-CBZ = extended-release carbamazepine capsules

Figure 1. Bipolar Subtype of Patient Population

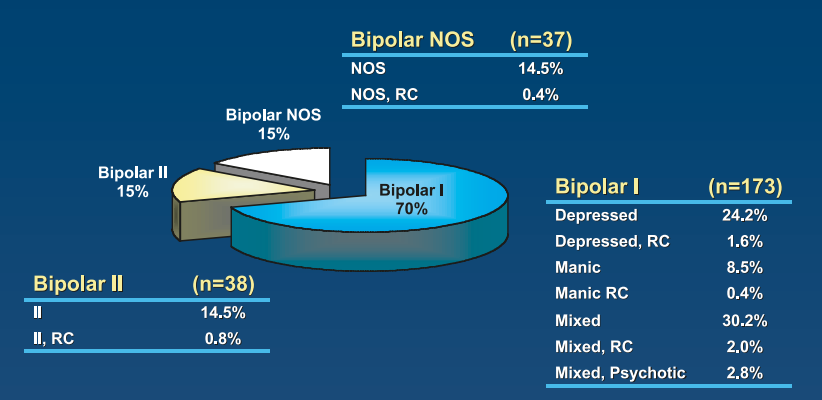

Bipolar NOS (n=37)
NOS 14.5%
NOS, RC 0.4%
Bipolar NOS 15%
Bipolar II 15%
Bipolar I 70%
Bipolar I (n=173)
Depressed 24.2%
Depressed, RC 1.6%
Manic 8.5%
Manic RC 0.4%
Mixed 30.2%
Mixed, RC 2.0%
Mixed, Psychotic 2.8%
Bipolar II (n=38)
II 14.5%
II, RC 0.8%

Data on file, Lawrence D. Ginsberg, MD, Red Oak Psychiatry Associates.
RC = rapid cycling; NOS = not otherwise specified

Figure 2. Severity of Illness (CGI-S) at ERC-CBZ Initiation

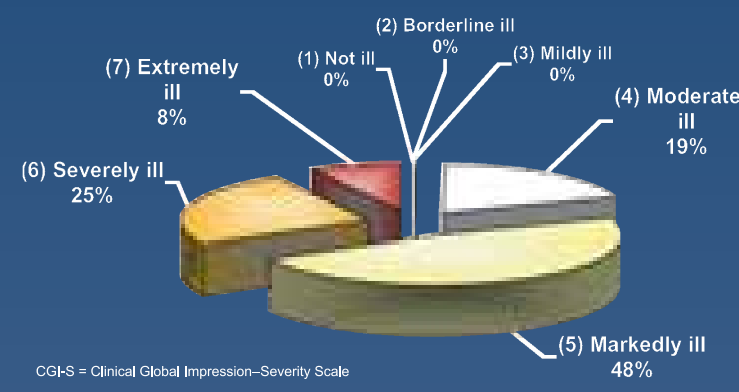

(2) Borderline ill 0%
(1) Not ill 0%
(3) Mildly ill 0%
(7) Extremely ill 8%
(4) Moderately ill 19%
(6) Severely ill 25%
(5) Markedly ill 48%

CGI-S = Clinical Global Impression–Severity Scale

Figure 3. Patient Response and Relapse on ERC-CBZ

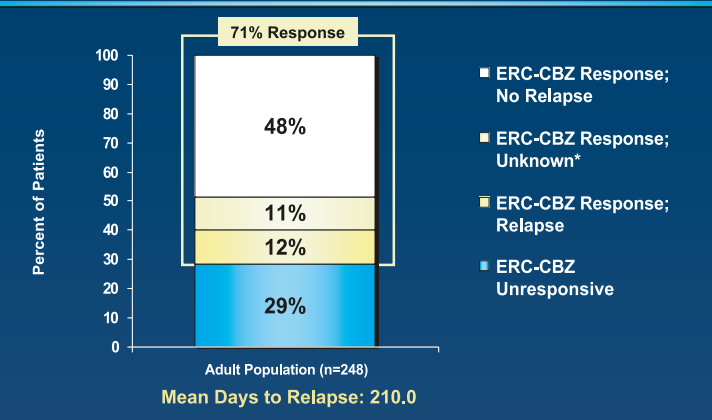

71% Response
Percent of Patients
48%
11%
12%
29%
Adult Population (n=248)
Mean Days to Relapse: 210.0
ERC-CBZ Response; No Relapse
ERC-CBZ Response; Unknown*
ERC-CBZ Response; Relapse
ERC-CBZ Unresponsive

ERC-CBZ response is defined as achieving a CGI-I ≤3. Relapse is defined by a change in CGI-I to ≥ 4 after an observed ERC-CBZ response.
*Response unknown includes: lost to follow-up, moved out of location; switched physicians, couldnít afford treatment, etc.
Data on file, Lawrence D. Ginsberg, MD, Red Oak Psychiatry Associates.

Figure 4. Analysis of ERC-CBZ Responders' CGI-I Scores

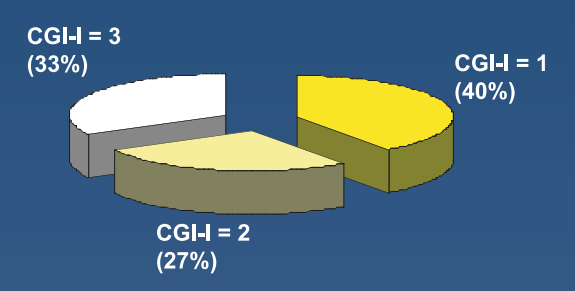

CGI-I = 3 (33%)
CGI-I = 1 (40%)
CGI-I = 2 (27%)

CGI-I = Clinical Global Impression – Improvement scale
ERC-CBZ = Extended-release carbamazepine capsules

Figure 5. Dosage Distribution of ERC-CBZ at Best CGI-I

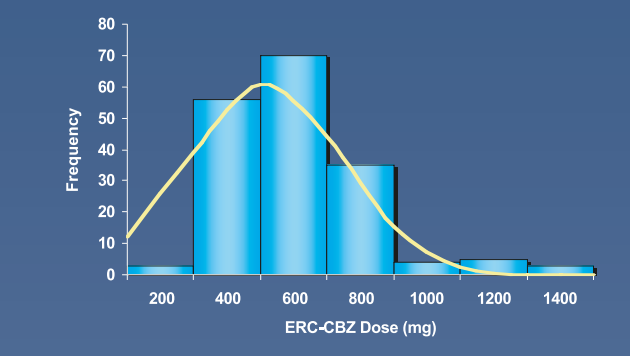

Frequency
ERC-CBZ Dose (mg)

CGI-I = Clinical Global Impression–Improvement Scale

RESULTS

- Of the 248 patients reviewed in this study, 71% were female, and ages ranged from 18–84 years, with a mean age of 35 years. Forty-nine percent of patients were initiated on ERC-CBZ monotherapy, while 6% of patients were able to be maintained on a once-daily dose of ERC-CBZ (Table 1)
- Seventy percent of patients were diagnosed with bipolar I disorder, 15% with bipolar II disorder, and 15% with bipolar disorder not otherwise specified (Figure 1)
- The severity of bipolar illness at ERC-CBZ initiation was determined using the CGI-S scale. Eighty-one percent of patients were determined to be at least markedly ill, scoring ≥ 5 on the CGI-S (Figure 2)
- One hundred seventy-six patients (71%) responded to ERC-CBZ
- Of the responders, 48% did not relapse, 11% were lost to follow-up, and 12% relapsed. The mean time to relapse was 210 days (95% CI, 97-323) (Figure 3)
- Analysis of ERC-CBZ responders revealed 40% of patients having achieved a CGI-I score of 1, 27% a score of 2, and 33% a score of 3 (Figure 4)
- At patients' best CGI-I score, the mean ERC-CBZ dose was 617 ± 231.4 mg/d, and the mean ERC-CBZ blood level was 7.1 ± 1.8 μg/mL (Figure 5; Table 1)
- Common treatment-emergent adverse events included somnolence (11.3%), nausea (8.1%), and dizziness (7.7%) (Table 2)
- Mean weight changed from 174.2 ± 44.4 lbs at ERC-CBZ initiation to 172.0 ± 40.6 lbs at drug discontinuation. Mean change was –3.2 ± 11.9 lbs (n=30 [smaller sample size due to missing data])
- There was no evidence of agranulocytosis or aplastic anemia

CONCLUSIONS

- This large retrospective study shows that ERC-CBZ appears to be effective in the treatment of bipolar disorder with a response rate similar to previous ERC-CBZ trials
- ERC-CBZ was effective in a broad spectrum of bipolar disorder
- Dosage and CBZ blood levels were similar to previous trials as was a low incidence of side effects, with no evidence of agranulocytosis or aplastic anemia, and no associated weight gain

REFERENCES

1. McElroy SL, Keck PE Jr. Pharmacologic agents for the treatment of acute bipolar mania. *Biol Psychiatry*. 2000;48:539-557.
2. Jensen PK, Moller A, Gram L, et al. Pharmacokinetic comparison of two carbamazepine slow-release formulations. *Acta Neurol Scand.* 1990;82:135-137.
3. Hoppener RJ, Kuyer A, Meijer JW, et al. Correlation between daily fluctuations of carbamazepine serum levels and intermittent side effects. *Epilepsia.* 1980;21:341-350.
4. Canger R, Altamura AC, Belvedere O, et al. Conventional vs controlled-release carbamazepine: a multicentre, double-blind, cross-over study. *Acta Neurol Scand.* 1990;82:9-13.
5. Haefeli WE, Meyer PG, Luscher TF. Circadian carbamazepine toxicity. *Epilepsia.* 1994;35:400-402.
6. Mirza WU, Rak IW, Thadani VM, et al. Six-month evaluation of Carbatrol (extended-release carbamazepine) in complex partial seizures. *Neurology.* 1998;51:1727-1729.
7. Weisler RH, Kalali AH, Ketter TA, and the SPD417 Study Group. A multicenter, randomized, double-blind, placebo-controlled trial of extended-release carbamazepine capsules as monotherapy for bipolar disorder patients with manic or mixed episodes. *J Clin Psychiatry.* In press.
8. Data on file, Shire Pharmaceutical Development. 2003.