ABSTRACT 314

Background: Carbamazepine extended-release capsules (CBZ-ERC) (Equetro™; Shire, Wayne, Pa) have been shown to possess significant efficacy in reducing symptoms of acute bipolar mania when administered using a twice-daily (bid) schedule. However, retrospective data suggest that once-daily CBZ-ERC dosing may have similar efficacy in the treatment of acute bipolar mania. To evaluate this hypothesis, a prospective trial comparing the efficacy of a standard bid dosing regimen with that of a once-nightly (qhs) CBZ-ERC regimen in patients with bipolar disorder was conducted.

Methods: The current 12-week, double-blind study involved adult outpatients experiencing either a manic or mixed bipolar episode at study entry. Trial participants were randomized to receive CBZ-ERC on either a bid or a qhs schedule. In both treatment groups, patients received a total CBZ-ERC dose of 200 to 1600 mg/d, with dose titration performed over the first 4 weeks postbaseline. Efficacy and safety were assessed at weeks 1, 2, 3, 4, 6, 8, and 12.

Results: In the intent-to-treat population (qhs, n = 53; bid, n = 52), least-squares mean changes in Young Mania Rating Scale scores relative to baseline showed no statistical difference between treatment arms at week 1 (bid, –5.3 vs qhs, –5.0; *P*=.841), and similar findings were made at all subsequent time points out to week 12 (bid, –11.4 vs qhs, –10.1; *P*=.325 [last observation carried forward {LOCF}]). The bid and qhs schedules also had comparable effects on overall disease severity (measured using the Clinical Global Impressions–Bipolar Disorder scale) at all time points from week 1 onward, with 50.0% of bid-treated patients and 41.5% of qhs-treated patients classified as "much improved" or "very much improved" at study end point (*P*=.534 [LOCF]). With regard to depressive symptoms, both treatment regimens yielded similar mean improvements in Hamilton Depression Rating Scale (*P*≥.287 [LOCF]) and Montgomery-Åsberg Depression Rating Scale scores (*P*≥.492 [LOCF]) at all postbaseline time points.

Conclusions: Once-nightly and bid dosing of CBZ-ERC appear to be similarly effective in reducing the severity of acute bipolar mania, as well as in reducing depressive symptoms and overall disease activity in patients with acute manic or mixed bipolar disorder. These findings suggest that qhs dosing of CBZ-ERC may represent an effective alternative to bid dosing for some patients; however, larger-scale double-blind trials are needed to fully confirm the validity of this hypothesis.

COMPARATIVE EFFICACY OF TWICE-DAILY AND ONCE-DAILY CARBAMAZEPINE EXTENDED-RELEASE CAPSULES IN BIPOLAR DISORDER: RESULTS FROM A DOUBLE-BLIND, PARALLEL-GROUP TRIAL

Richard H. Weisler, MD, Duke University Medical Center, Durham, North Carolina, and the University of North Carolina, Chapel Hill, North Carolina; **Lawrence Ginsberg, MD,** Red Oak Psychiatry Associates, Houston, Texas; **Thomas Gazda, MD,** St Luke's Medical Center, Scottsdale, Arizona; **Joseph Kerkering, MBA,** Shire, Wayne, Pennsylvania

BACKGROUND

- Carbamazepine extended-release capsules (CBZ-ERC) (Equetro™; Shire, Wayne, Pa) are approved by the US Food and Drug Administration for use in the treatment of acute manic or mixed episodes associated with Bipolar I Disorder
- Phase 3 clinical trials have demonstrated that CBZ-ERC possesses significant efficacy in the treatment of acute manic and mixed bipolar symptoms when it is administered according to a twice-daily (bid) dosing schedule[1,2]
- Although CBZ-ERC is approved for use according to a bid treatment schedule, anecdotal reports indicate that clinicians often prescribe regimens in which CBZ-ERC is taken once nightly (qhs) for the treatment of bipolar symptoms. Furthermore, findings from a recent retrospective chart study suggest that once-daily and bid dosing schedules exhibit comparable efficacy and safety profiles[3]
- In an effort to characterize prospectively the appropriateness of once-daily CBZ-ERC treatment regimens for patients with Bipolar I Disorder, a 12-week, randomized, double-blind, parallel-group clinical trial comparing qhs dosing of CBZ-ERC with bid dosing was conducted

METHODS

- Following a 2- to 10-day screening period during which eligibility for study entry was evaluated, adult outpatients with Bipolar I Disorder (most recent episode manic or mixed; Young Mania Rating Scale [YMRS] score ≥16 at screening) were randomized to receive treatment with CBZ-ERC on either a qhs or a bid dosing schedule **(Figure 1)**
- In both treatment arms, CBZ-ERC therapy was started at a total daily dose of 200 mg. Over the first 4 weeks of the study, total daily doses were titrated upward or downward in 200-mg steps every 3 to 4 days as necessary to achieve optimal efficacy and tolerability (allowed daily dose range, 200-1600 mg; target daily dose, 800 mg), after which patients continued to receive their identified optimal CBZ-ERC dose for the remainder of the study
 - To maintain blinding, each patient received treatment from a blister card that was divided into an "AM" section (containing unlabeled capsules to be taken during the morning) and a "PM" section (containing unlabeled capsules to be taken at night)
 - Blister cards issued to the qhs group contained only placebo capsules in the AM section and only CBZ-ERC capsules in the PM section
 - Blister cards issued to the bid group contained half of each day's CBZ-ERC dose in the AM section and the remaining half of each day's CBZ-ERC dose in the PM section
 - The 4-week dose titration schedule used in the current outpatient trial differs from the more rapid 1-week titration schedule that was used in previous CBZ-ERC trials,[1,2] which involved inpatients with acute manic or mixed symptoms
- Evaluations of treatment efficacy, safety, and tolerability were performed once weekly during the initial 4-week dose titration period and then at weeks 6, 8, and 12
 - Instruments used to assess treatment efficacy included the YMRS (primary efficacy measure), the Clinical Global Impressions–Bipolar Disorder scale (CGI-BP), the Hamilton Depression Rating Scale (HDRS), and the Montgomery-Åsberg Depression Rating Scale (MADRS)
 - Patients who withdrew from the study before its completion were assessed for efficacy, safety, and tolerability on their last day of treatment or as soon as possible (≤2 days) thereafter
- In addition to being assessed for efficacy, safety, and tolerability during the course of study treatment, all patients underwent a final evaluation of treatment safety and tolerability (via telephone or in-person interview) 30 days after receiving their last dose of study drug

RESULTS

Patient Characteristics, Patient Disposition, and Drug Dosing

- The current trial enrolled a total of 111 patients (mean age, 37.0 years), 69 of whom were women (62.2%) and 85 of whom were white (76.6%). There were no noteworthy differences between the qhs treatment arm (n = 58) and the bid treatment arm (n = 53) with regard to demographics or disease characteristics at baseline **(Table)**
 - Of the 111 patients enrolled in the current study, 105 (qhs, n = 53; bid, n = 52) were included in the intent-to-treat (ITT) population, the primary population for efficacy evaluation. Patients included in the ITT population were those who had 1) been randomized, 2) received at least 1 dose of the study drug, 3) undergone a baseline YMRS score evaluation, and 4) had at least 1 postbaseline YMRS score evaluation

Figure 1. Study Design

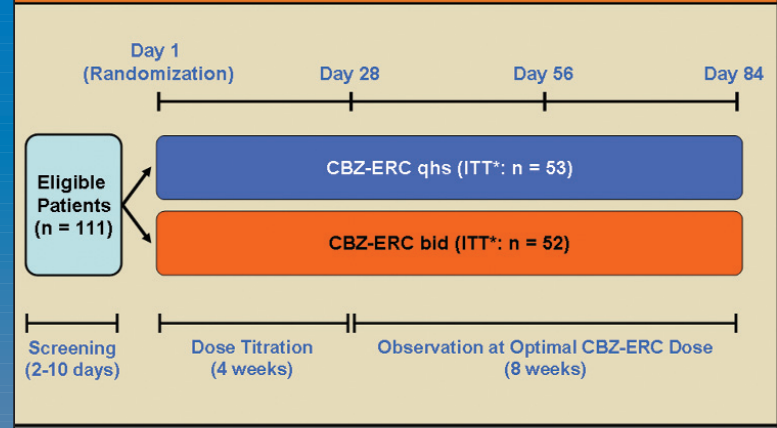

Day 1 (Randomization)
Day 28
Day 56
Day 84
Eligible Patients (n = 111)
CBZ-ERC qhs (ITT*: n = 53)
CBZ-ERC bid (ITT*: n = 52)
Screening (2-10 days)
Dose Titration (4 weeks)
Observation at Optimal CBZ-ERC Dose (8 weeks)

*Includes all patients who received ≥1 dose of study drug and had Young Mania Rating Scale scores evaluated at baseline and at ≥1 postbaseline time point.
qhs = once nightly; bid = twice daily; ITT = intent-to-treat; CBZ-ERC = carbamazepine extended-release capsules.

Figure 2. Changes in Manic Symptoms in Association With qhs and bid Dosing of CBZ-ERC

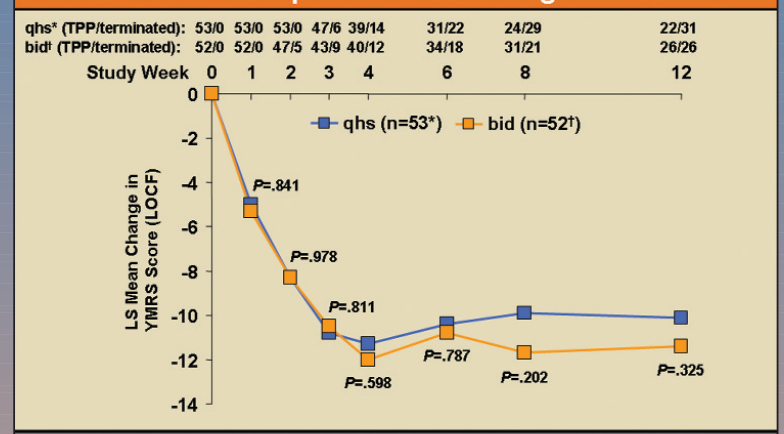

qhs* (TPP/terminated): 53/0 53/0 53/0 47/6 39/14 31/22 24/29 22/31
bid† (TPP/terminated): 52/0 52/0 47/5 43/9 40/12 34/18 31/21 26/26
Study Week 0 1 2 3 4 6 8 12
qhs (n=53*) bid (n=52†)
LS Mean Change in YMRS Score (LOCF)
P=.841
P=.978
P=.811
P=.598
P=.787
P=.202
P=.325

*ITT population. Five additional patients were randomized but withdrew before meeting the criteria for inclusion in the ITT analysis.
†ITT population. One additional patient was randomized but withdrew before meeting the criteria for inclusion in the ITT analysis.
ITT = intention-to-treat; TPP = treated per protocol; qhs = once nightly; bid = twice daily; CBZ-ERC = carbamazepine extended-release capsules; LS = least-squares; YMRS = Young Mania Rating Scale; LOCF = last observation carried forward.

Figure 3. Changes in Depressive Symptoms in Association With qhs and bid Dosing of CBZ-ERC

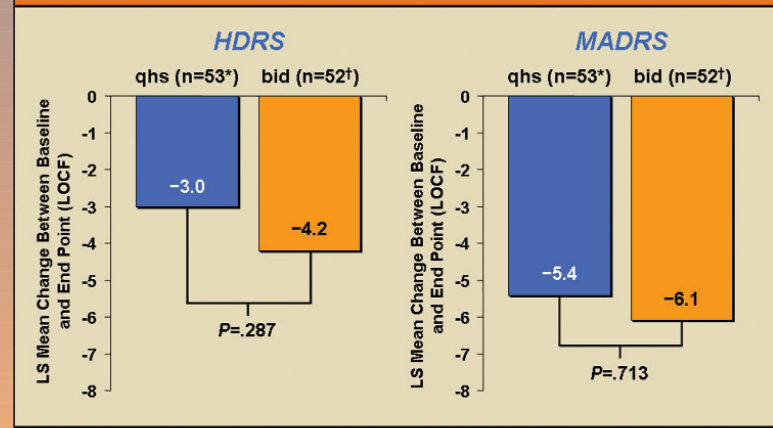

HDRS
MADRS
qhs (n=53*) bid (n=52†)
LS Mean Change Between Baseline and End Point (LOCF)
−3.0
−4.2
P=.287
−5.4
−6.1
P=.713

*LOCF population includes 16 patients who completed the entire course of study treatment and 37 patients who did not.
†LOCF population includes 25 patients who completed the entire course of study treatment and 27 patients who did not.
qhs = once nightly; bid = twice daily; CBZ-ERC = carbamazepine extended-release capsules; HDRS = Hamilton Depression Rating Scale; MADRS = Montgomery-Åsberg Depression Rating Scale; LS = least-squares; LOCF = last observation carried forward.

Figure 4. Disease Remission in Association With qhs and bid Dosing of CBZ-ERC

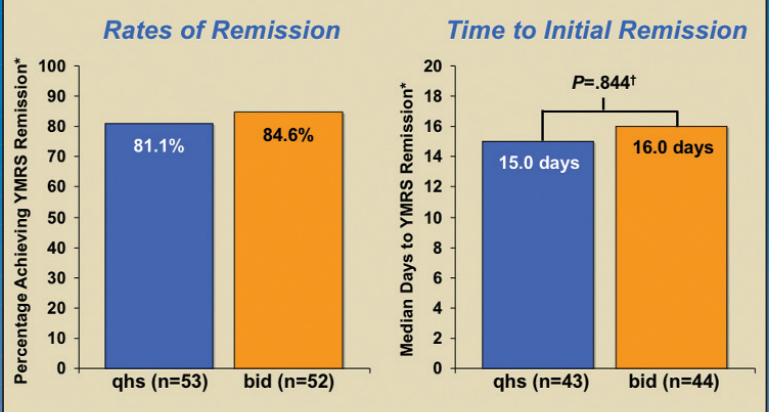

Rates of Remission
Time to Initial Remission
Percentage Achieving YMRS Remission*
81.1%
84.6%
qhs (n=53)
bid (n=52)
Median Days to YMRS Remission*
P=.844†
15.0 days
16.0 days
qhs (n=43)
bid (n=44)

*YMRS remission was defined as the attainment of a YMRS score ≤12.
† Log-rank test.
qhs = once nightly; bid = twice daily; CBZ-ERC = carbamazepine extended-release capsules; YMRS = Young Mania Rating Scale

Figure 5. Global Symptom Responses to qhs and bid Treatment With CBZ-ERC

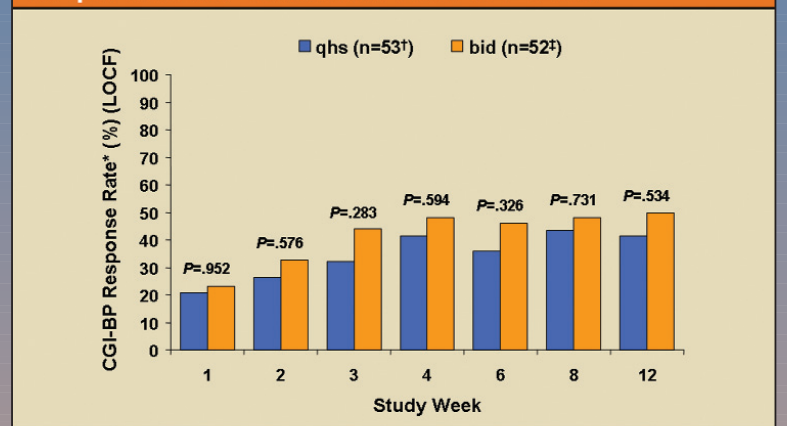

qhs (n=53†) bid (n=52‡)
CGI-BP Response Rate* (%) (LOCF)
P=.952
P=.576
P=.283
P=.594
P=.326
P=.731
P=.534
Study Week

*Patients judged to be "much improved" or "very much improved" with regard to overall disease severity according to the CGI-BP were classified as responders.
†LOCF population includes 16 patients who completed the entire course of study treatment and 37 patients who did not.
‡LOCF population includes 25 patients who completed the entire course of study treatment and 27 patients who did not.
qhs = once nightly; bid = twice daily; CBZ-ERC = carbamazepine extended-release capsules;
CGI-BP = Clinical Global Impressions–Bipolar Disorder scale; LOCF = last observation carried forward.

Table. Baseline Patient Characteristics

Characteristic	Treated qhs	Treated bid
Enrolled, N	58	53
Gender, n (%)		
Male	19 (32.8%)	23 (43.4%)
Female	39 (67.2%)	30 (56.6%)
Race/Ethnicity, n (%)		
White	44 (75.9%)	41 (77.4%)
Black	8 (13.8%)	6 (11.3%)
Hispanic	6 (10.3%)	6 (11.3%)
Mean YMRS total score ± SD	20.6 ± 6.6	22.8 ± 7.3
Mean HDRS total score ± SD	13.7 ± 6.9	13.6 ± 7.2
Mean MADRS score ± SD	17.8 ± 9.4	18.9 ± 10.5

qhs = once nightly; bid = twice daily; YMRS = Young Mania Rating Scale; HDRS = Hamilton Depression Rating Scale; MADRS = Montgomery-Åsberg Depression Rating Scale; SD = standard deviation.

RESULTS (CONT)

- A total of 64 patients (57.7% of all those randomized)—37 in the qhs treatment arm (63.8%) and 27 in the bid treatment arm (50.9%)—withdrew from the study before its completion
- The mean daily CBZ-ERC doses received by patients in the qhs and bid treatment arms were 656 mg and 727 mg, respectively
 - In both treatment arms, the most commonly administered daily CBZ-ERC dose was 800 mg

Treatment Efficacy

- At each postbaseline time point, last-observation-carried forward (LOCF) analysis revealed that there was no statistically significant difference between qhs-treated patients and bid-treated patients with regard to the observed least-squares (LS) mean change in YMRS scores relative to baseline **(Figure 2)**
 - The LS mean change in YMRS score between baseline and final evaluation was –10.1 points for patients in the qhs treatment arm, compared with –11.4 points over the same time period for patients in the bid treatment arm (*P*=.325)
- The qhs and bid dosing schedules were also associated with statistically comparable mean improvements (relative to baseline) in scores on the HDRS and the MADRS at all postbaseline time points (LOCF analysis) **(Figure 3)**
 - Patients in the qhs treatment arm experienced an LS mean improvement (ie, decrease) of 3.0 points in HDRS scores between baseline and final evaluation, while patients in the bid treatment arm experienced an LS mean improvement of 4.2 points over the same interval (*P*=.287)
 - With regard to MADRS scores, LS mean improvements (ie, decreases) of 5.4 points and 6.1 points relative to baseline were documented in the qhs and bid treatment arms, respectively, at final evaluation (*P*=.713)
- Disease remission (defined as the attainment of a YMRS score ≤12) was achieved by 81.1% of all qhs-treated patients and by 84.6% of all bid-treated patients. There was no significant difference between the 2 treatment arms in terms of the median time to first remission (qhs, 15.0 days vs bid, 16.0 days; *P*=.844) **(Figure 4)**
- The percentage of patients classified as responders (ie, patients who were "much improved" or "very much improved" relative to baseline) in terms of overall disease severity according to the CGI-BP generally increased over the course of the study, with response rates in the qhs and bid treatment arms statistically comparable at all postbaseline time points **(Figure 5)**
 - At final evaluation, 41.5% of patients in the qhs treatment arm, compared with 50.0% of patients in the bid treatment arm, met the criteria for response according to the CGI-BP (*P*=.534)

CONCLUSIONS

- Over the course of the current 12-week, randomized, double-blind, parallel-group study involving patients with manic or mixed bipolar symptoms, qhs and bid CBZ-ERC dosing schedules were not associated with significantly different mean changes in YMRS, HDRS, or MADRS scores or CGI-BP response rates at any postbaseline time point. In addition, there was no significant difference between qhs dosing and bid dosing in terms of median time to disease remission
- Treatment with CBZ-ERC, whether administered according to a qhs or a bid dosing schedule, was associated with absolute reductions in the severity of manic and depressive symptoms, as well as with decreased global disease severity, in patients with bipolar disorder
- In light of the observed statistical equivalence of the 2 CBZ-ERC dosing schedules examined in the current study with regard to therapeutic efficacy, it appears that qhs dosing of CBZ-ERC may serve as an appropriate alternative to bid dosing in the treatment of patients with bipolar disorder. However, further investigation geared toward characterizing the potential advantages or any disadvantages associated with qhs dosing of this extended-release formulation is desirable

Equetro is a trademark of Shire LLC.

REFERENCES

1. Weisler RH, Kalali AH, Ketter TA. A multicenter, randomized, double-blind, placebo-controlled trial of extended-release carbamazepine capsules as monotherapy for bipolar disorder patients with manic or mixed episodes. *J Clin Psychiatry.* 2004;65:478-484.
2. Weisler RH, Keck PE Jr, Swann AC, Cutler AJ, Ketter TA, Kalali AH. Extended-release carbamazepine capsules as monotherapy for acute mania in bipolar disorder: a multicenter, randomized, double-blind, placebo-controlled trial. *J Clin Psychiatry.* 2005;66:323-330.
3. Ginsberg LD. Safety and efficacy of carbamazepine extended-release capsules in patients with bipolar disorder: QD vs BID. *Ann Clin Psychiatry.* 2006;18(suppl 1):27-30.

Supported by funding from Shire.

Presented at the 46th Annual Meeting of the New Clinical Drug Evaluation Unit; June 12-15, 2006; Boca Raton, Florida.